Poster number:

330

Innate defense regulators are a novel macrophage-mediated therapy to treat infectious and inflammatory diseases

Shunsuke Takenaka [1], Annett Rozek [1], Agnieszka Kielczewska [1], Lisa Thorson [2], John R. North [1], B. Brett Finlay [2], Oreola Donini [1]

[1] Inimex Pharmaceuticals Inc, Burnaby, British Columbia, Canada

[2] Department of Microbiology and Immunology, University of British Columbia, Vancouver, British Columbia, Canada

Background

Innate Defense Regulators (IDRs) are a novel class of synthetic peptides with no antimicrobial activity that enhance microbial infection control while suppressing inflammation. Treatment of mice with IDRs enhanced their survival in bacterial infection models and reduced bacterial burden. The anti-infective effect of IDRs was maintained in animals depleted of neutrophils, but abolished in animals depleted of macrophages/monocytes, indicating an important role for macrophages in IDR mediated host protection. IDRs have been shown to increase chemokine levels at the site of infection (CCL5 and CXCL10), enhance macrophage recruitment to the site of infection and improve the resolution of bacterial infection alone or in conjunction with antibiotics. IDRs have also been shown to modulate pathogen-free inflammation, where levels of pro-inflammatory cytokines were also reduced. Collectively, these data suggest that IDRs modulate innate immune responses by enhancing macrophage recruitment while suppressing inflammatory pathways. The recent identification of the intracellular target of IDR, p62 (sequestosome-1), which is ubiquitously present in the body and known to modulate inflammatory pathways in macrophages, suggests further possibilities for the therapeutic application of IDRs. Recently a Phase 1 study with a lead IDR, IMX942, has demonstrated the safety of this novel, first-in-class, therapeutic approach.

Results

IDRs protect animals from infection

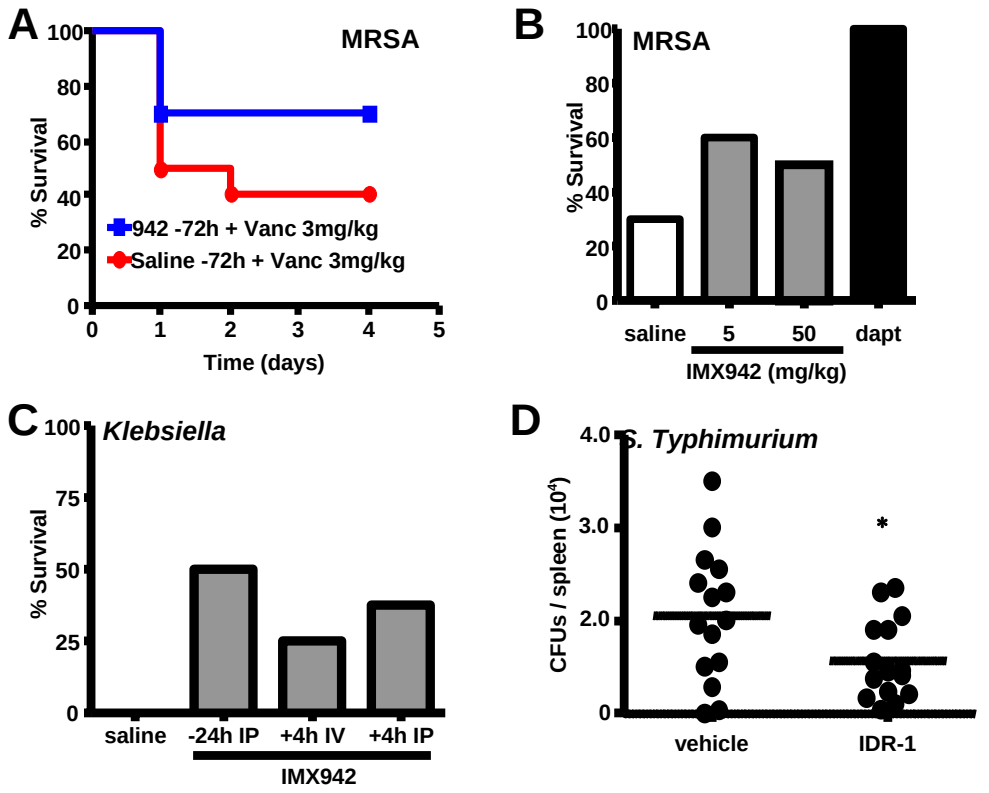
A
MRSA
% Survival
942 -72h + Vanc 3mg/kg
Saline -72h + Vanc 3mg/kg
Time (days)
B
MRSA
% Survival
saline
5
50
dapt
IMX942 (mg/kg)
C
Klebsiella
% Survival
saline
-24h IP
+4h IV
+4h IP
IMX942
D
S. Typhimurium
CFUs / spleen (10⁴)
vehicle
IDR-1

Figure 1. A. Female CF-1 mice (n = 10/group) were treated with IMX942 (50 mg/kg IV) or saline 72 hours prior to infection with MRSA (UC6685, 8.2 x 10^7 CFU/mouse IP). Vancomycin (3 mg/kg SC) was administered 1 and 5 hours after infection. **B.** CD-1 mice were treated with IMX942 (5 or 50 mg/kg IV) or daptomycin 4 hours prior to infection with MRSA (8.4 x 10^7 CFU/mouse IP) and their survival was monitored for 4 days. **C.** Female Balb/c mice (n = 8/group) were treated with 24 mg/kg of IMX942 either 24 hours prior to (IP) or 4 hours post (IP or IV) infection with *Klebsiella* (2.8 x 10^5 CFU/mouse IP) and their survival was determined at 24 hours after infection. **D.** Balb/c mice (n = 15/group) were treated with IDR-1 or vehicle control and infected with *S. Typhimurium* SL1344 (1 $\times$ 10^4 CFU/mouse IP) 4 hours later. Bacterial CFU counts were determined for the spleen of individual mice after 24 hours of infection.

IDRs recruit macrophages *in vivo*

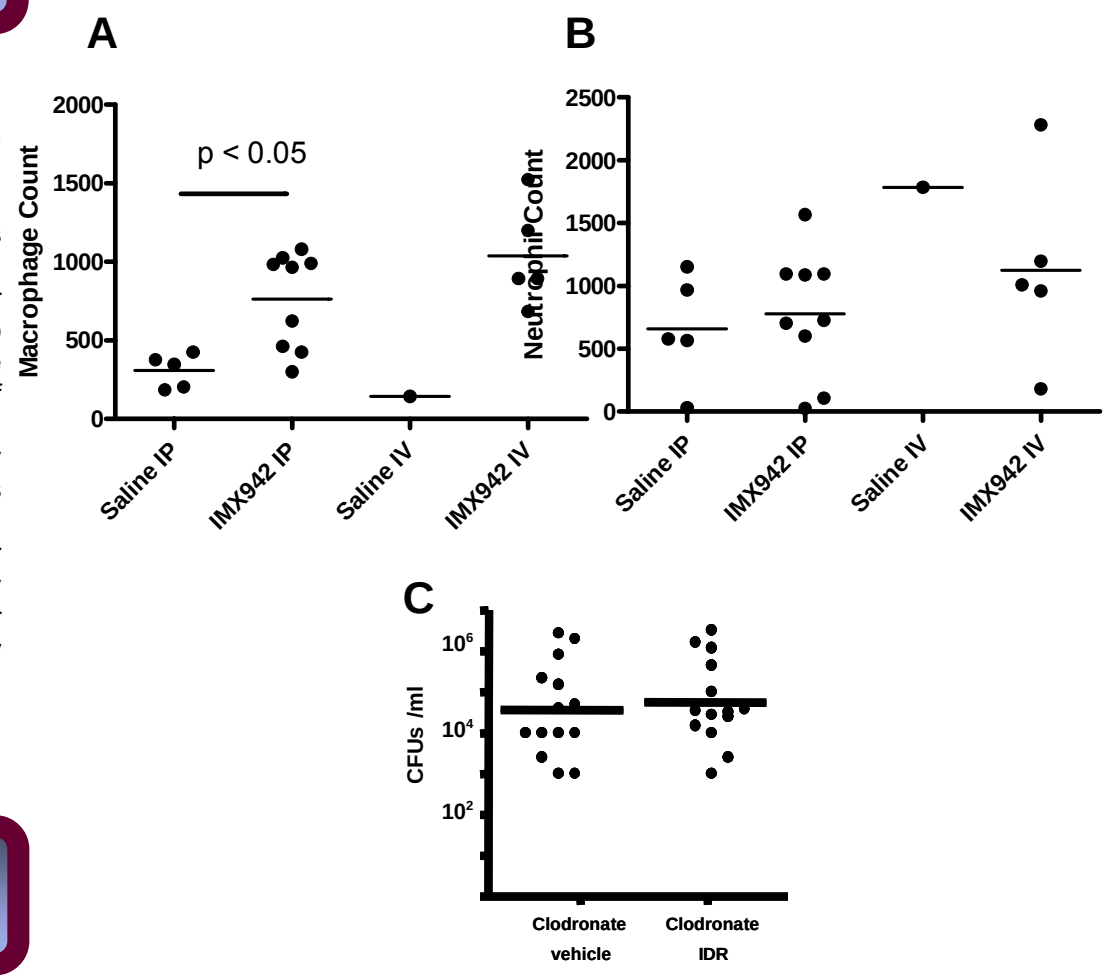
A
p < 0.05
Macrophage Count
Saline IP
IMX942 IP
Saline IV
IMX942 IV
B
Neutrophil Count
Saline IP
IMX942 IP
Saline IV
IMX942 IV
C
CFUs /ml
Clodronate vehicle
Clodronate IDR

Figure 2. Importance of macropahges on IDR mediated protection from bacterial infection. Animals were infected IP with *S. aureus* and treated with IMX942 or control saline either IP or IV 4 hours after the infection. Peritoneal lavage was collected 24 hours infection and cell numbers were determined by cytospin. **A.** Macrophage numbers. **B.** Neutrophil numbers. **C**. Effect of liposomal clodronate depletion of macrophages on peritoneal bacterial clearance.

IDRs protect leukopenic animals

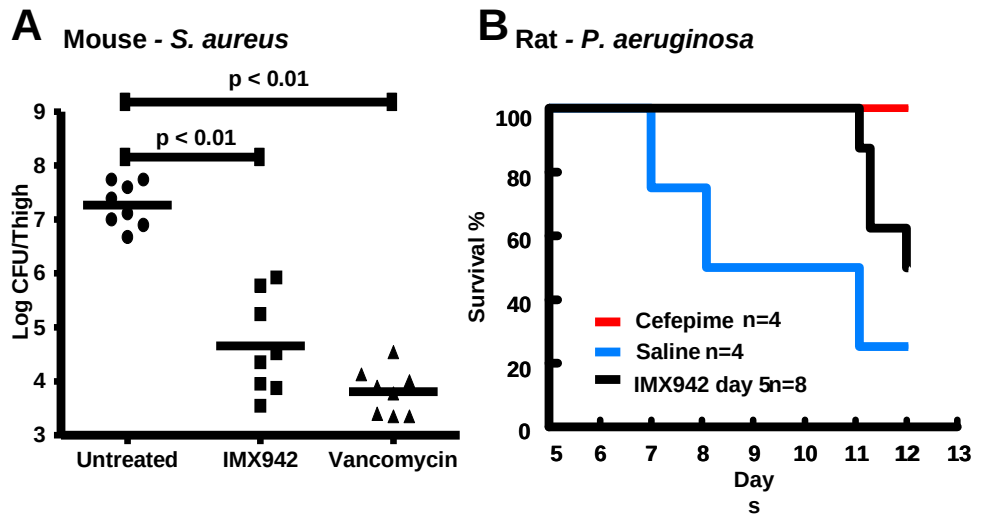
A
Mouse - S. aureus
p < 0.01
p < 0.01
Log CFU/Thigh
Untreated
IMX942
Vancomycin
B
Rat - P. aeruginosa
Survival %
Cefepime n=4
Saline n=4
IMX942 day 5n=8
Days

Figure 3. A. Cyclophosphamide treated female swiss albino mice were treated with IMX942 (50 mg/kg IM) 24h prior to infection with *S. aureus* (~9.5 x 10^5 cfu/mouse IM). Vancomycin (100 mg/kg SC) was administered at 1, 6 and 18h after infection. CFU in the infected thigh was examined 24h post infection. **B.** Female Sprague-Dawley rates were treated IM with cefamandole 4 days prior to infection to disrupt GI flora and also treated with cyclophosphamide to induce leukopenia. Rats were infected with oral *P. aeruginosa* (1 x 10^6 CFU/mL) on days 0, 2 and 4. On day 5, animals received IMX942 (10 mg/kg IV), cefepime (40 mg/kg IM for 3 days) or saline (IM for 3 days) their survival was monitored.

IDRs modulates cytokines/chemokines *in vivo*

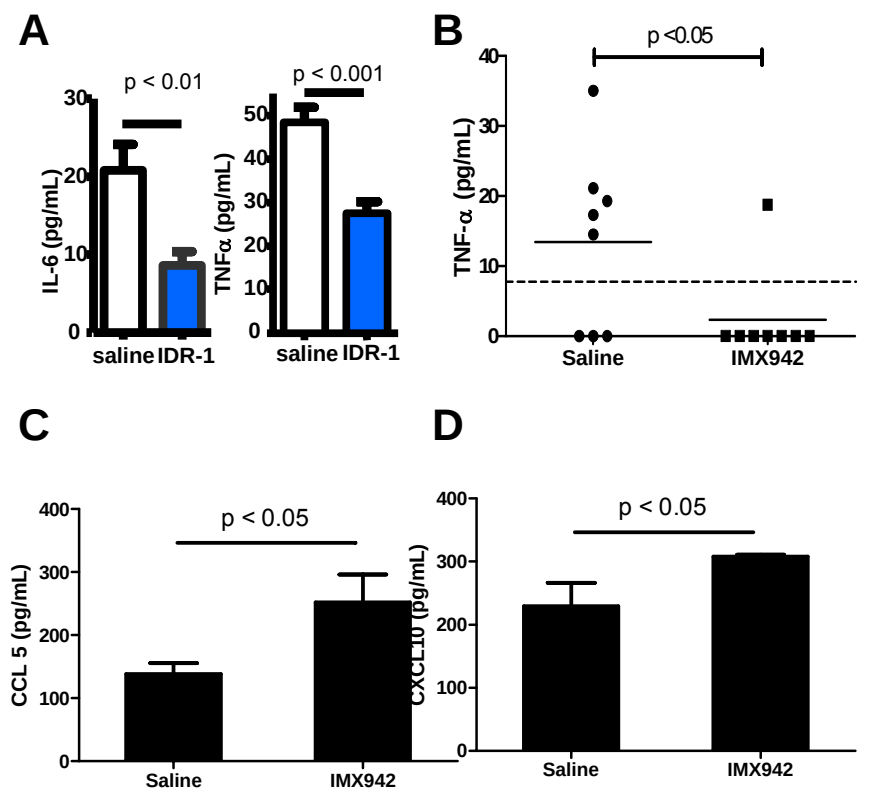
A
p < 0.01
p < 0.001
IL-6 (pg/mL)
TNFα (pg/mL)
saline
IDR-1
B
p <0.05
TNF-α (pg/mL)
Saline
IMX942
C
p < 0.05
CCL 5 (pg/mL)
Saline
IMX942
D
p < 0.05
CXCL10 (pg/mL)
Saline
IMX942

Figure 5. A. IL-6 and TNF-α levels in spleen homogenate from female Balb/c mice (n=15/group) treated with IDR-1 (24 mg/kg IP) or vehicle control and infected with *S. Typhimurium* SL1344 (1 $\times$ 10^4 CFU/mouse IP) 4h later. Spleens collected 24h post infection **B.** TNF-α level in the peritoneal lavage from female CD-1 mice (n=8/group) infected with *S. aureus* (1.8 $\times$ 10^6 CFU/mouse IP). Mice were treated with IMX942 (30 mg/kg IP) or saline 24h prior to infection. Peritoneal lavage collected 24h post-infection. **C and D.** CCL5 and CXCL10 levels in the same infection model as Figure 5B. Peritoneal lavage samples collected 3h post-infection.

IDR-1 protects animals from cerebral malaria

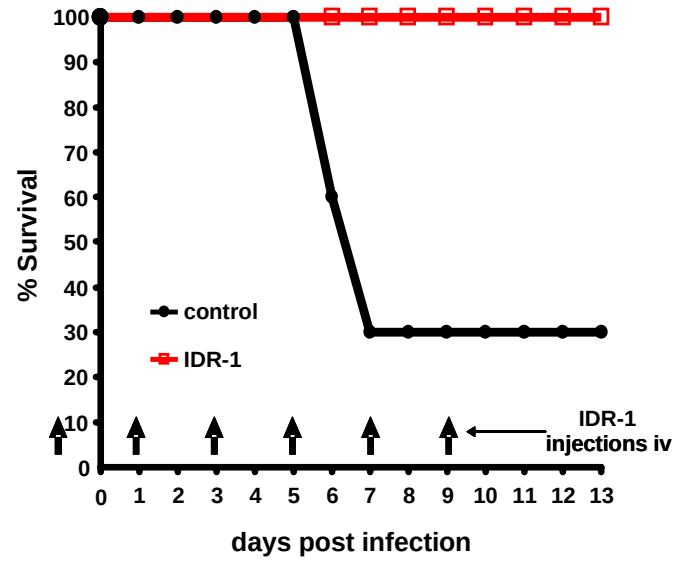
% Survival
control
IDR-1
IDR-1 injections iv
days post infection

Figure 6. Female C57BL/6 mice were infected with 1 x 10^6 *P. berghei* ANKA parasites IP on Day 0. Animals were treated with IDR-1 (24 mg/kg IV) or control saline on days -1, 1, 3, 5, 7 and 9 and their survival was monitored.

IDR-1 modulates inflammation *in vivo*

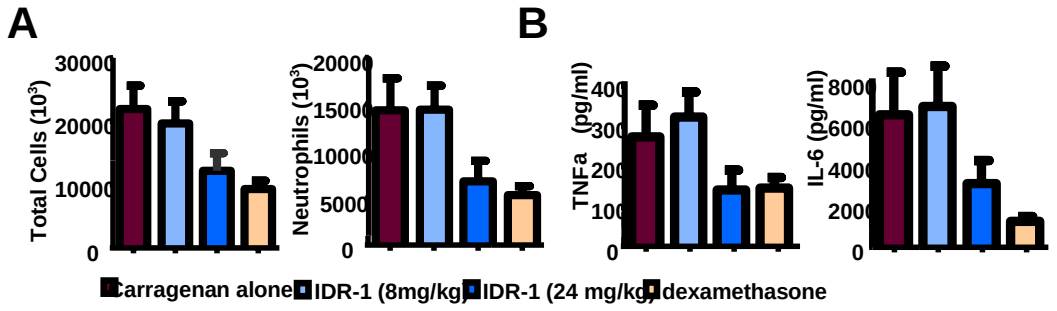
A
Total Cells (10³)
Neutrophils (10³)
B
TNFa (pg/ml)
IL-6 (pg/ml)
Carragenan alone
IDR-1 (8mg/kg)
IDR-1 (24 mg/kg)
dexamethasone

Figure 6. IDR-1 reduces the inflammatory response in a mouse air pouch model of acute inflammation. Animals were injected with carrageenan solution to induce inflammation. IDR-1 was administered 12 hr prior to carrageenan treatment. Control mice were either left untreated (carrageenan alone) or pre-treated with dexamethasone (5 mg/kg). **A.** Number of total leukocytes and neutrophils in the air pouch. **B.** TNF-α and IL-6 levels in the air pouch lavage fluid.

IMX942 shifts inflammatory status in healthy human

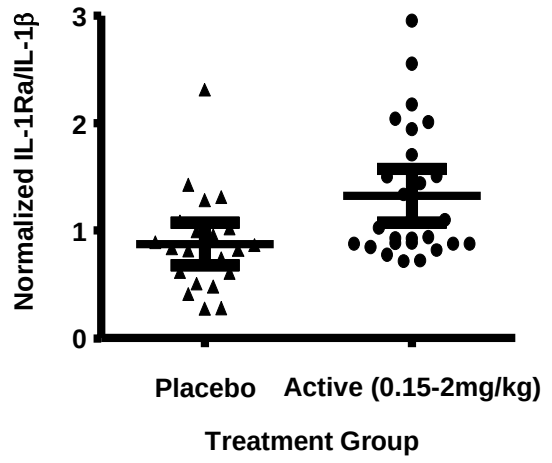
Normalized IL-1Ra/IL-1β
Placebo
Active (0.15-2mg/kg)
Treatment Group

Figure 7. Healthy volunteers were treated with a single injection of IMX942 (0.15 – 2 mg/kg IV, n = 26) or placebo (n = 22) and blood samples were collected 1h post treatment. Whole blood samples were then stimulated with LPS *in vitro* for 4 hours and IL-1Ra and IL-1β levels in the plasma were examined. Individual data were normalized against pre-treatment values and presented as IL-1Ra/IL-1β ratio.

Conclusions

- IDRs target host to provide protection from bacterial infection
- Macropahges, but not neutrophils, are required for IDR-mediated host protection
- IDRs modulate inflammatory responses
- IMX942 is well tolerated by human
- IMX942 shifts inflammatory status in human