SGX942 REDUCES THE DURATION OF SEVERE ORAL MUCOSITIS

O. Donini[1], M. Kudrimoti[2], A. Curtis[3], S. Azawi[4], F. Worden[5], S. Katz[6], J. Elder[7]

[1]Soligenix, Inc [2]University of Kentucky [3]Gibbs Cancer Center [4]VA Long Beach Hospital [5]University of Michigan Health System [6]Willis-Knighton Cancer Center [7]PharPoint Research

INTRODUCTION

SGX942 (dusquetide[1,2]) is a first-in-class Innate Defense Regulator[3, 4] with a novel mechanism of action, modulating the innate immune system to decrease inflammation and enhance bacterial clearance and tissue healing. Chemoradiation (CRT) associated OM has been linked to dysfunctional inflammation stimulated by innate immunity.

Head and neck cancer (HNC) patients receiving CRT are at high risk for ulcerative (WHO score ≥2) and severe (WHO score ≥3) OM. Severe OM (SOM) is associated with an inability to eat and/or drink, and may result in dehydration, infection, the need for parenteral nutrition, hospitalization and the interruption of CRT. It ~~has a~~ adversely affects quality of life for patients and has a significant pharmacoeconomic impact.

.

STUDY DESIGN

A double blind, placebo-controlled Phase 2 exploratory study in 111 HNC patients receiving at least 55 Gy radiation and cisplatin (weekly: 30-40 mg/m^2; every 3rd week: 80-100 mg/m^2) was conducted. Subjects were treated with study drug (placebo or SGX942 at 1.5 or 6.0 mg/kg) twice weekly with a 4-minute IV infusion. A 3.0 mg/kg dose group was included for dose escalation safety purposes *only*. OM (WHO score) was monitored twice weekly until end of radiation and then weekly for one month.

Follow-up visits are ongoing.

CONSORT Table

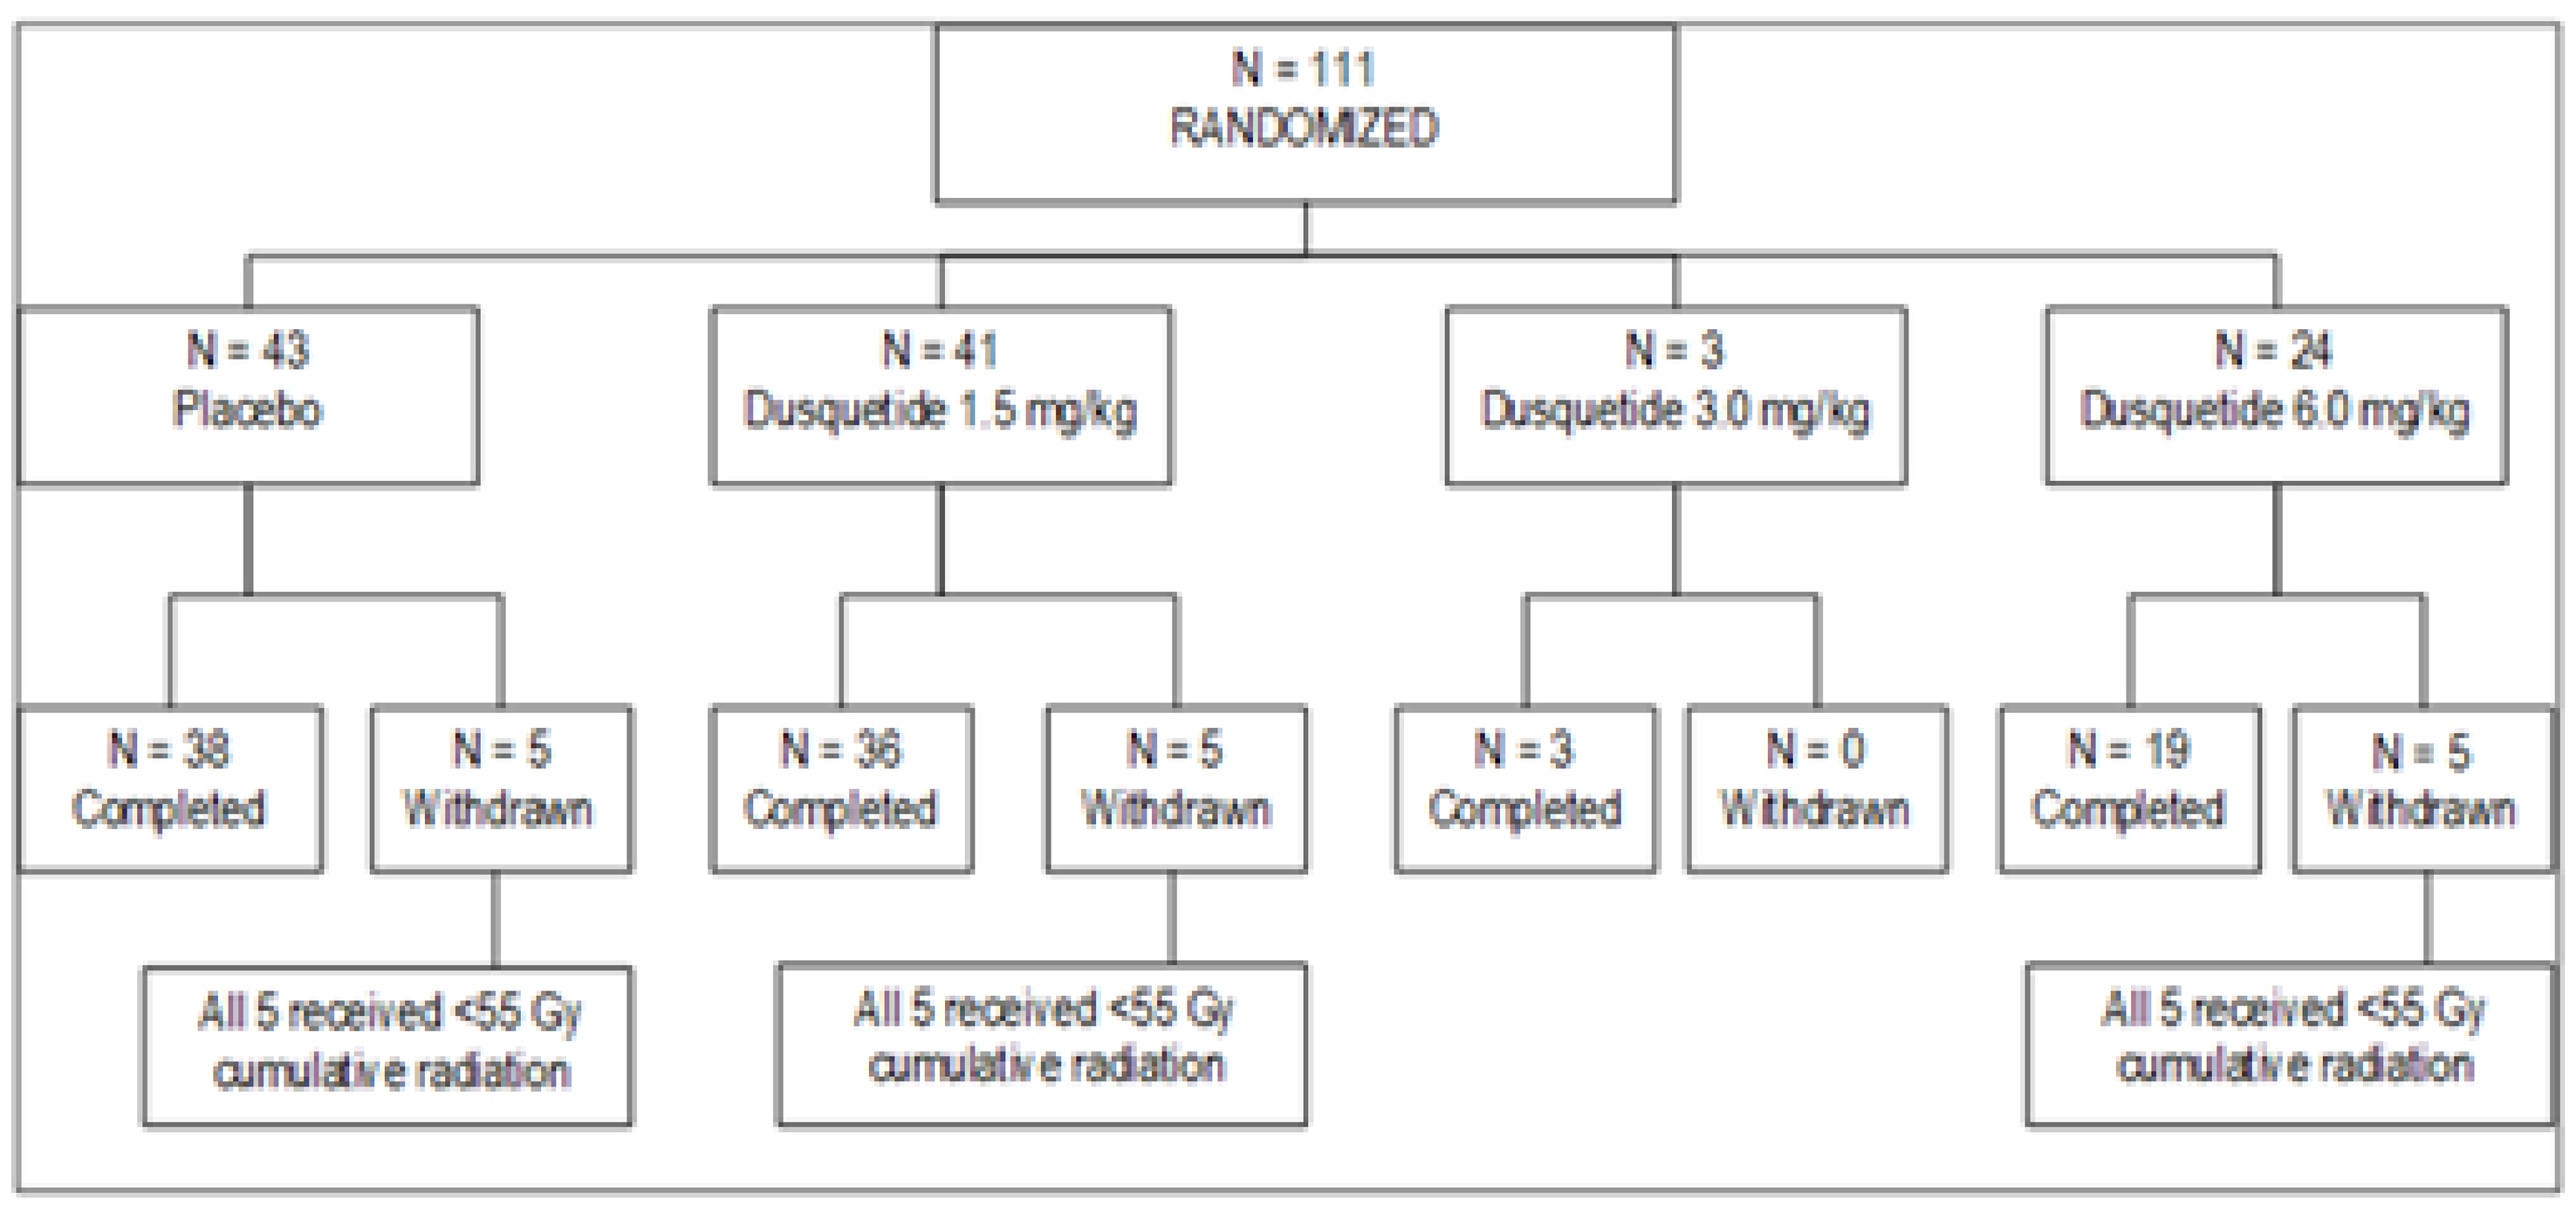
N = 111
RANDOMIZED
N = 43
Placebo
N = 41
Dusquetide 1.5 mg/kg
N = 3
Dusquetide 3.0 mg/kg
N = 24
Dusquetide 6.0 mg/kg
N = 38
Completed
N = 5
Withdrawn
N = 36
Completed
N = 5
Withdrawn
N = 3
Completed
N = 0
Withdrawn
N = 19
Completed
N = 5
Withdrawn
All 5 received <55 Gy cumulative radiation
All 5 received <55 Gy cumulative radiation
All 5 received <55 Gy cumulative radiation

SAFETY

No treatment emergent changes in vital signs, laboratory values, adverse events or serious adverse events were detected. In this patient population, SGX942 was safe and well-tolerated, consistent with the findings of the Phase 1 clinical study in 84 healthy volunteers.

Tumor status at the one-month follow-up visit favored the 1.5 mg/kg SGX942 treatment group.

Non-fungal infections were also decreased in the SGX942 treated groups, consistent with the preclinical data.

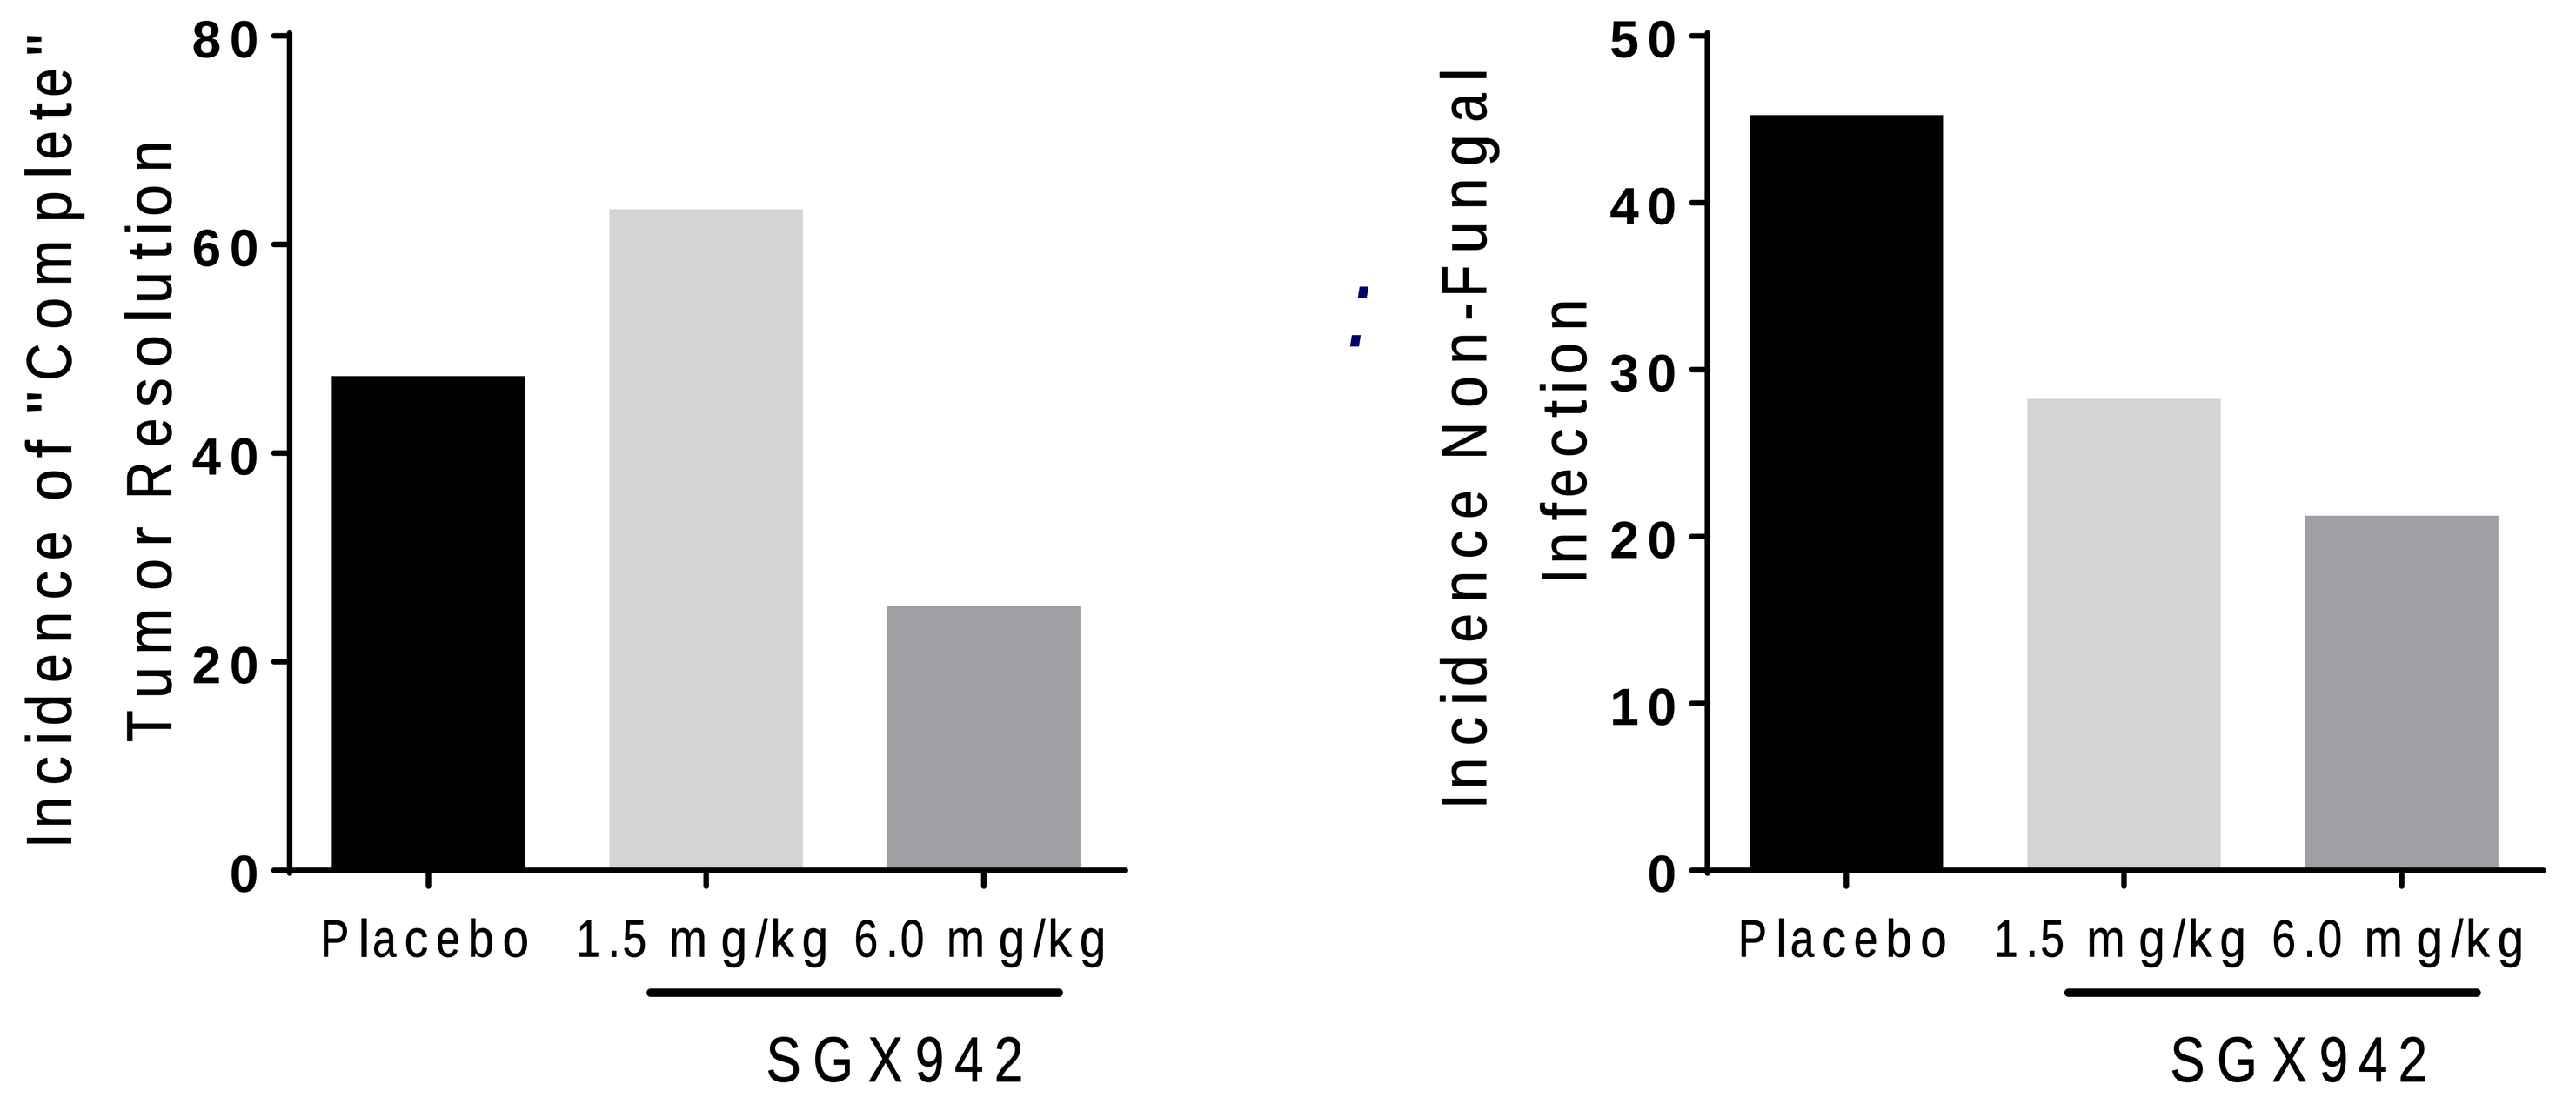
Incidence of "Complete" Tumor Resolution
80
60
40
20
0
Placebo
1.5 mg/kg
6.0 mg/kg
SGX942
Incidence Non-Fungal Infection
50
40
30
20
10
0
Placebo
1.5 mg/kg
6.0 mg/kg
SGX942

EFFICACY

- In this exploratory Phase 2 study, statistical significance was prospectively defined as p<0.10. Duration was defined as the elapsed days between the first occurrence of OM and the last recorded occurrence of OM. Patients with no SOM were censored (duration = 0.1).
- Both ulcerative OM (UOM) and SOM were evaluated and the largest impact observed in SOM.
- Findings with the 1.5 mg/kg group were consistent across endpoints (incidence SOM, # days with SOM, onset SOM) and subpopulations (HPV+, HPV-, chemotherapy regimen, etc.). Results with the 6.0 mg/kg were not consistent. In the Phase 1 trial, a similar non-linear dose response curve was observed with inflammatory markers.

.

Duration of Severe Oral Mucositis

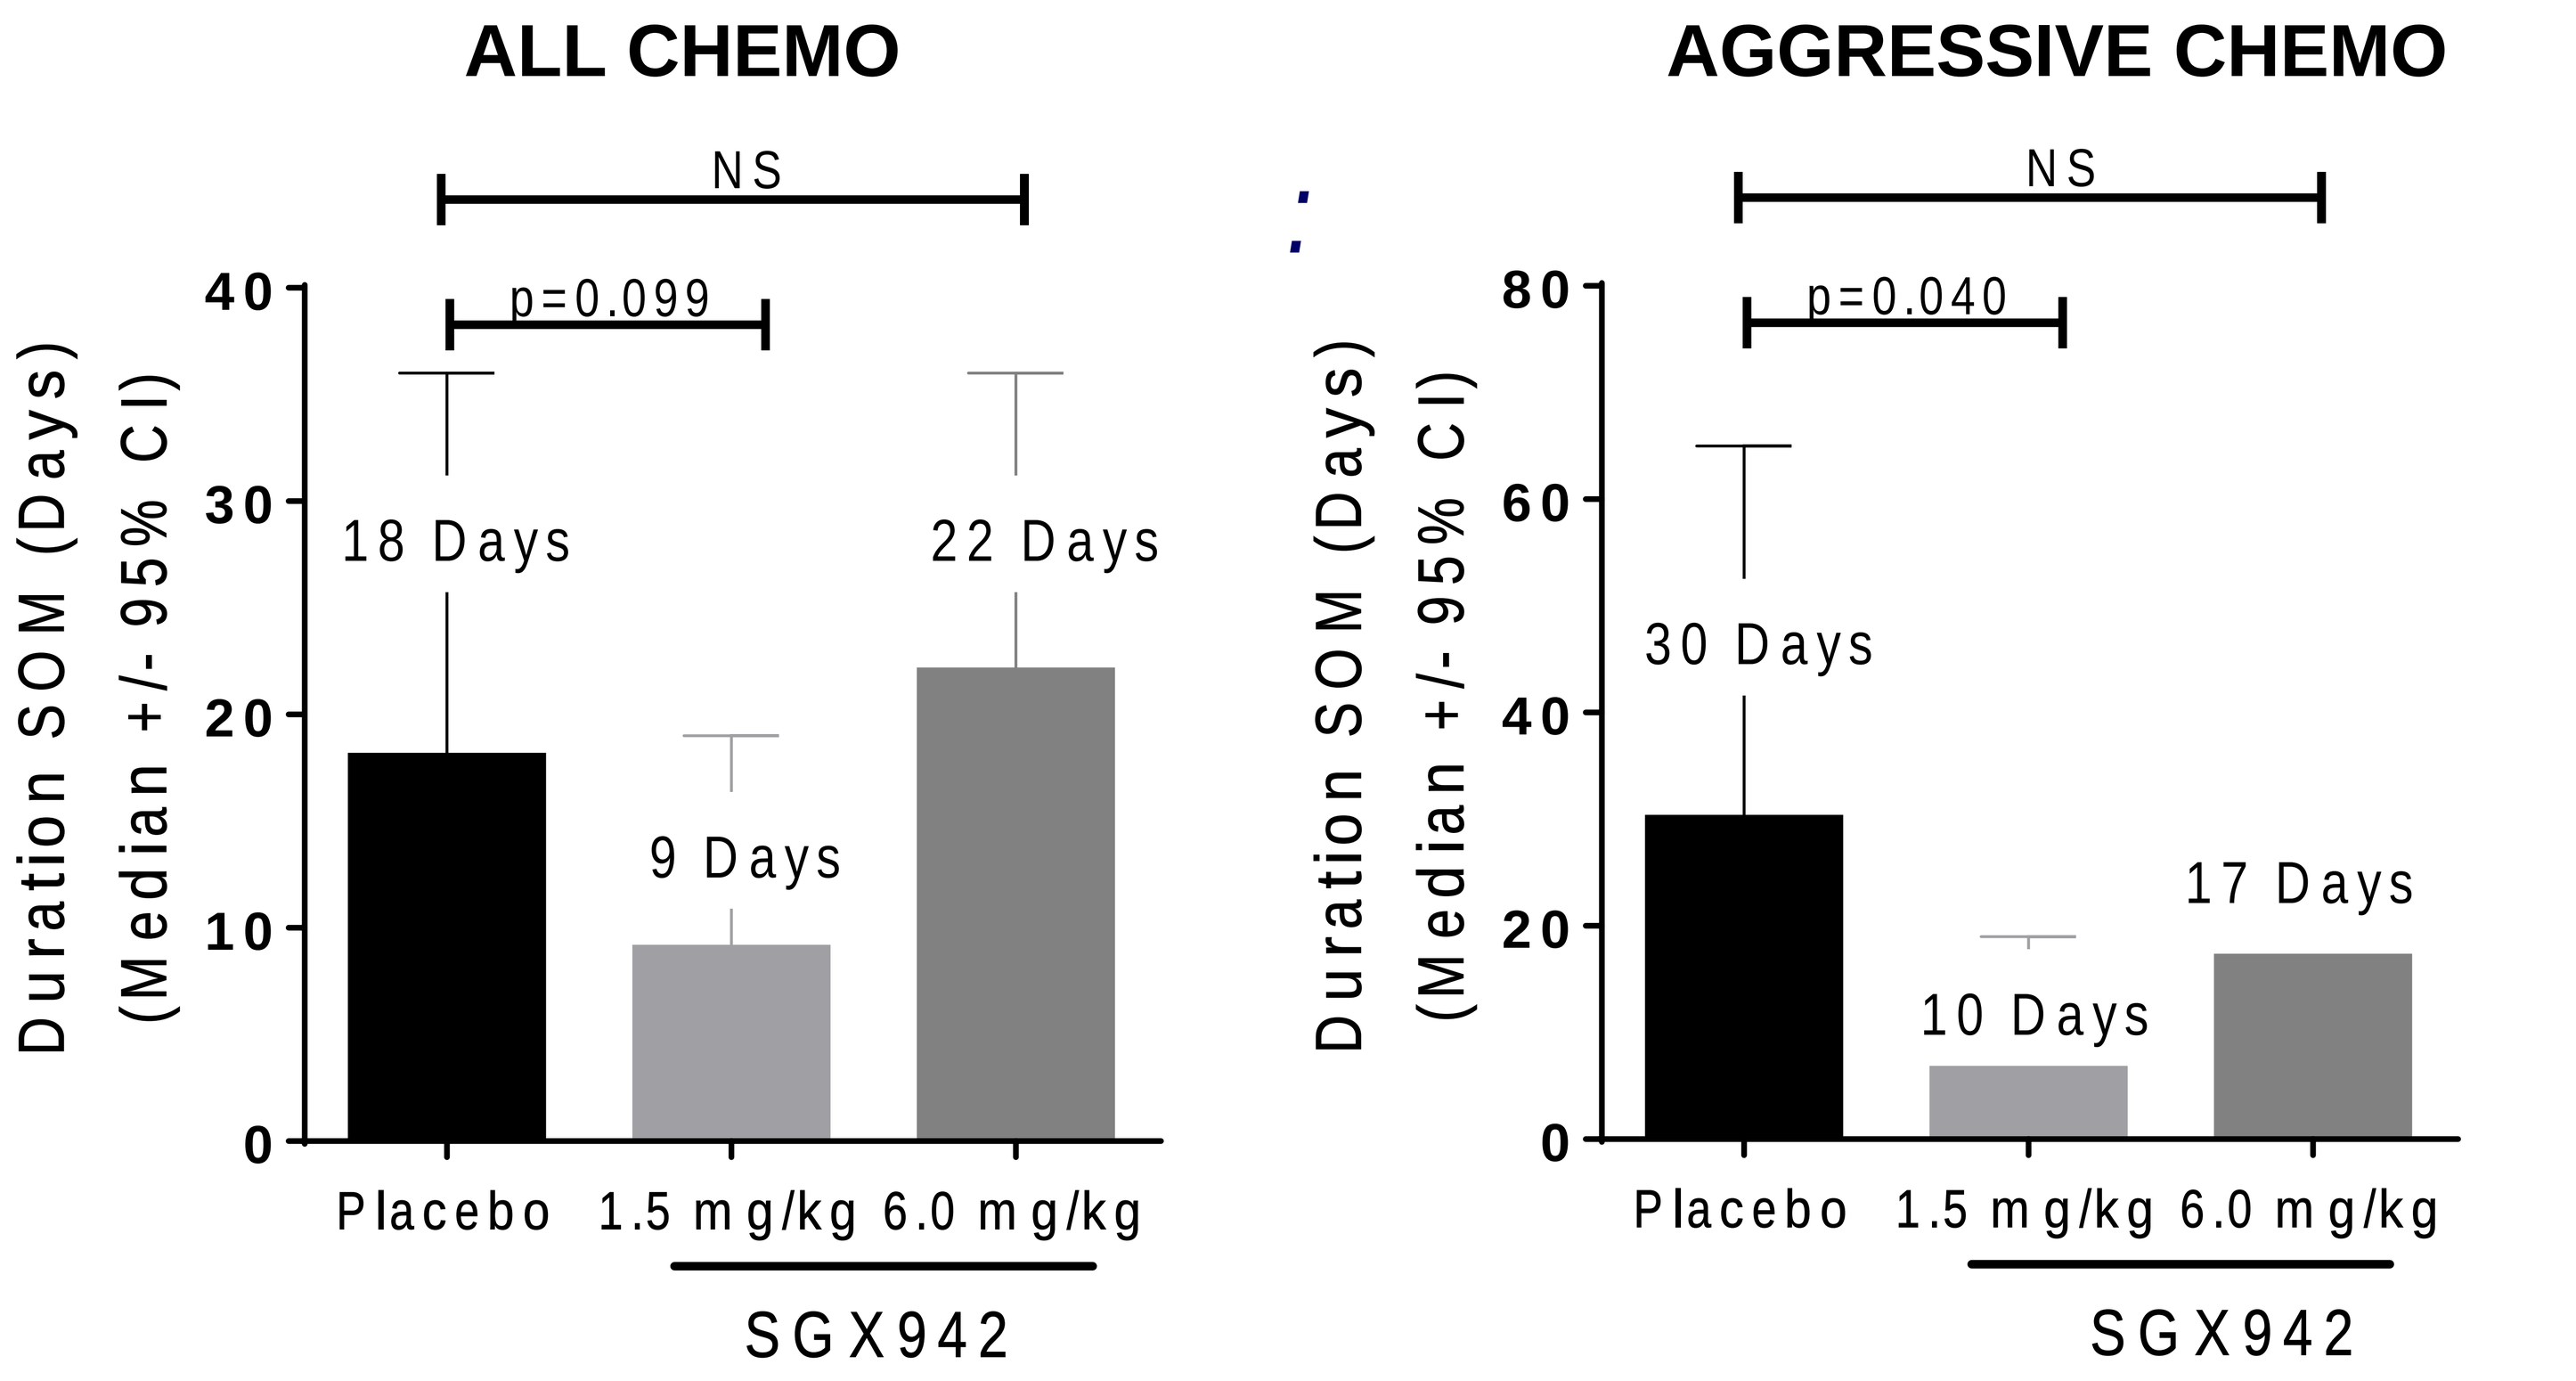
ALL CHEMO
NS
p=0.099
Duration SOM (Days) (Median +/- 95% CI)
40
30
20
10
0
18 Days
9 Days
22 Days
Placebo
1.5 mg/kg
6.0 mg/kg
SGX942
AGGRESSIVE CHEMO
NS
p=0.040
Duration SOM (Days) (Median +/- 95% CI)
80
60
40
20
0
30 Days
10 Days
17 Days
Placebo
1.5 mg/kg
6.0 mg/kg
SGX942

Anti-Inflammatory Activity in Phase 1 Clinical Study

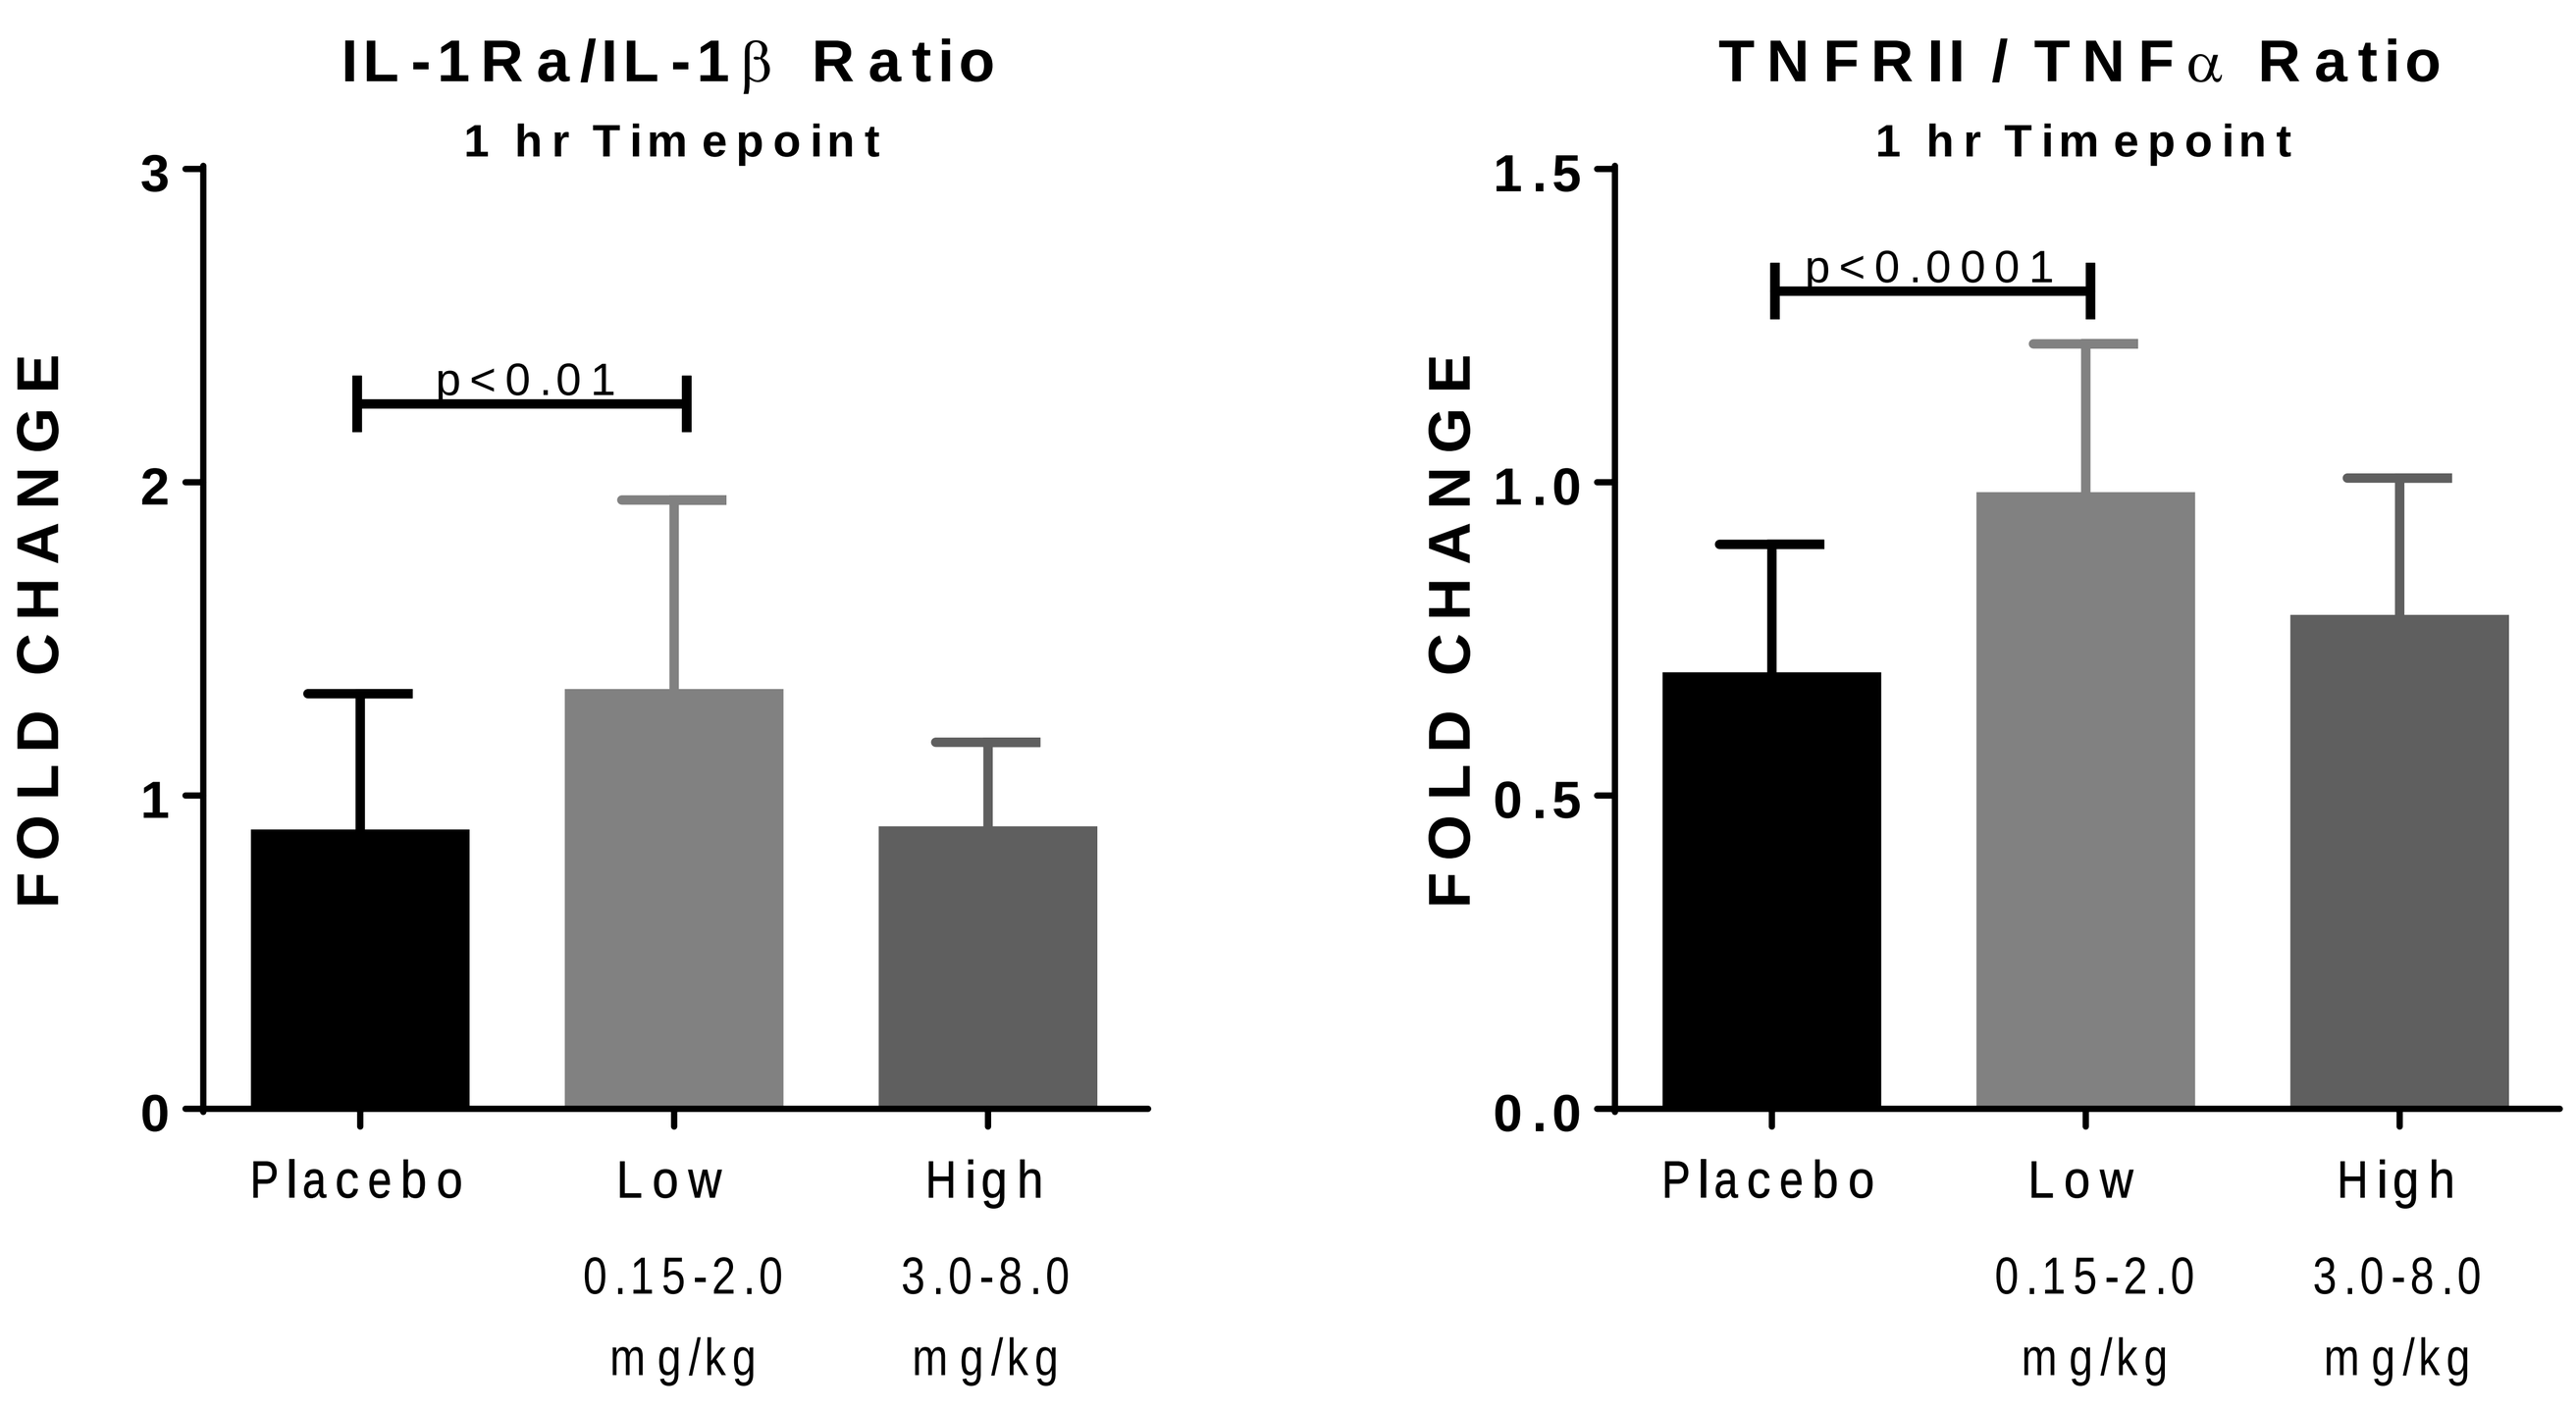
IL-1Ra/IL-1β Ratio
1 hr Timepoint
FOLD CHANGE
3
2
1
0
p<0.01
Placebo
Low
0.15-2.0 mg/kg
High
3.0-8.0 mg/kg
TNFRII / TNFα Ratio
1 hr Timepoint
FOLD CHANGE
1.5
1.0
0.5
0.0
p<0.0001
Placebo
Low
0.15-2.0 mg/kg
High
3.0-8.0 mg/kg

CONCLUSIONS

- A non-linear dose response curve was observed, consistent with Phase 1 clinical and preclinical studies.
- SGX942 (1.5 mg/kg) decreased SOM 50% and 67% in patients at highest risk for SOM.
- SGX942 (6.0 mg/kg) was less effective treating OM, consistent with previous clinical and preclinical data.
- Consistent with preclinical work, SGX942 reduced incidence of infection.
- SGX942 did not interfere with tumor treatment, also consistent with preclinical xenograft studies
- SGX942 was safe and well-tolerated in HNC cancer patients.

REFERENCES

1. 1. North et al. 2016. A novel approach for emerging and antibiotic resistant infections: Innate defense regulators as an agnostic therapy. *J. Biotech.* **226**: 24-34. http://dx.doi.org/10.1016/j.jbiotec.2016.03.032.
2. 2.Donini et al. 2010. Reduced Infection and Mucositis in Chemotherapy-Treated Animals Following Innate Defense Modulation Using a Novel Drug Candidate. *Blood (ASH Annual Meeting Abstracts)* 116:3781. http://soligenix.com/wp-content/uploads/poster_imx942_in_mucositis_and_infection.pdf
3. 3. Yu et al. 2009. Sequestosome-1/p62 is the key intracellular target of innate defense regulator peptide. *J Biol Chem.* 284:36007-11. http://www.jbc.org/content/284/52/36007.long
4. 4. Scott et al. 2007. An anti-infective peptide that selectively modulates the innate immune response. *Nat. Biotech.* 25: 465-72 . http://www.nature.com/nbt/journal/v25/n4/full/nbt1288.html